#142

Pharmacokinetics of a Single Dose of Sufentanil Sublingual Tablet 30 mcg vs. Intravenous Sufenta® 30 mcg in Healthy Volunteers

Sandra K. Willsie, DO, MA[1], Karen DiDonato, MSN, RN[2], Pamela P. Palmer, MD, PhD[2]

[2]PRA Health Sciences, Lenexa, KS; [2]AcelRx Pharmaceuticals Inc. Medical Affairs, Redwood City, CA

Introduction

Following completion of a comprehensive late-phase clinical program, a New Drug Application for the Sufentanil Sublingual Tablet 30 mcg (SST 30 mcg; DSUVIA™) was recently submitted to the FDA. The proposed indication for SST 30 mcg is management of moderate-to-severe acute pain in a medically-supervised setting such as the emergency department or out-patient surgery.

Sufentanil is a potent mu opioid agonist that exerts both sedative and analgesic effects. Currently it is approved for intravenous (IV) or epidural administration.[1] The onset of action of IV sufentanil is short, within 1 to 4 minutes after bolus administration, but the duration of effect is quite limited due to extensive first-pass metabolism of this lipophilic drug. U.S. physicians have over three decades of experience administering sufentanil via direct routes, recent clinical data suggests however that delivering the drug sublingually may provide analgesic advantages for some patients by facilitating a longer time period from peak plasma concentration (C_{max}) to 50% of C_{max} (context sensitive half-time, [$CST_{1/2}$]).[2-3] Additionally, sublingual administration of these small tablets does not trigger a salivary response, thereby allowing for more consistent mucosal uptake and more predictable timing to peak plasma concentrations.[4]

The objective of this analysis was to establish the single-dose and multiple-dose pharmacokinetics (PK) of sublingual administration of SST 30 mcg in addition to comparing the PK of a single dose of SST 30 mcg to IV sufentanil (Sufenta®; 50 mcg/mL) 30mcg infused over 1 minute.

Figure 1. The Sufentanil Sublingual Tablet 30 mcg

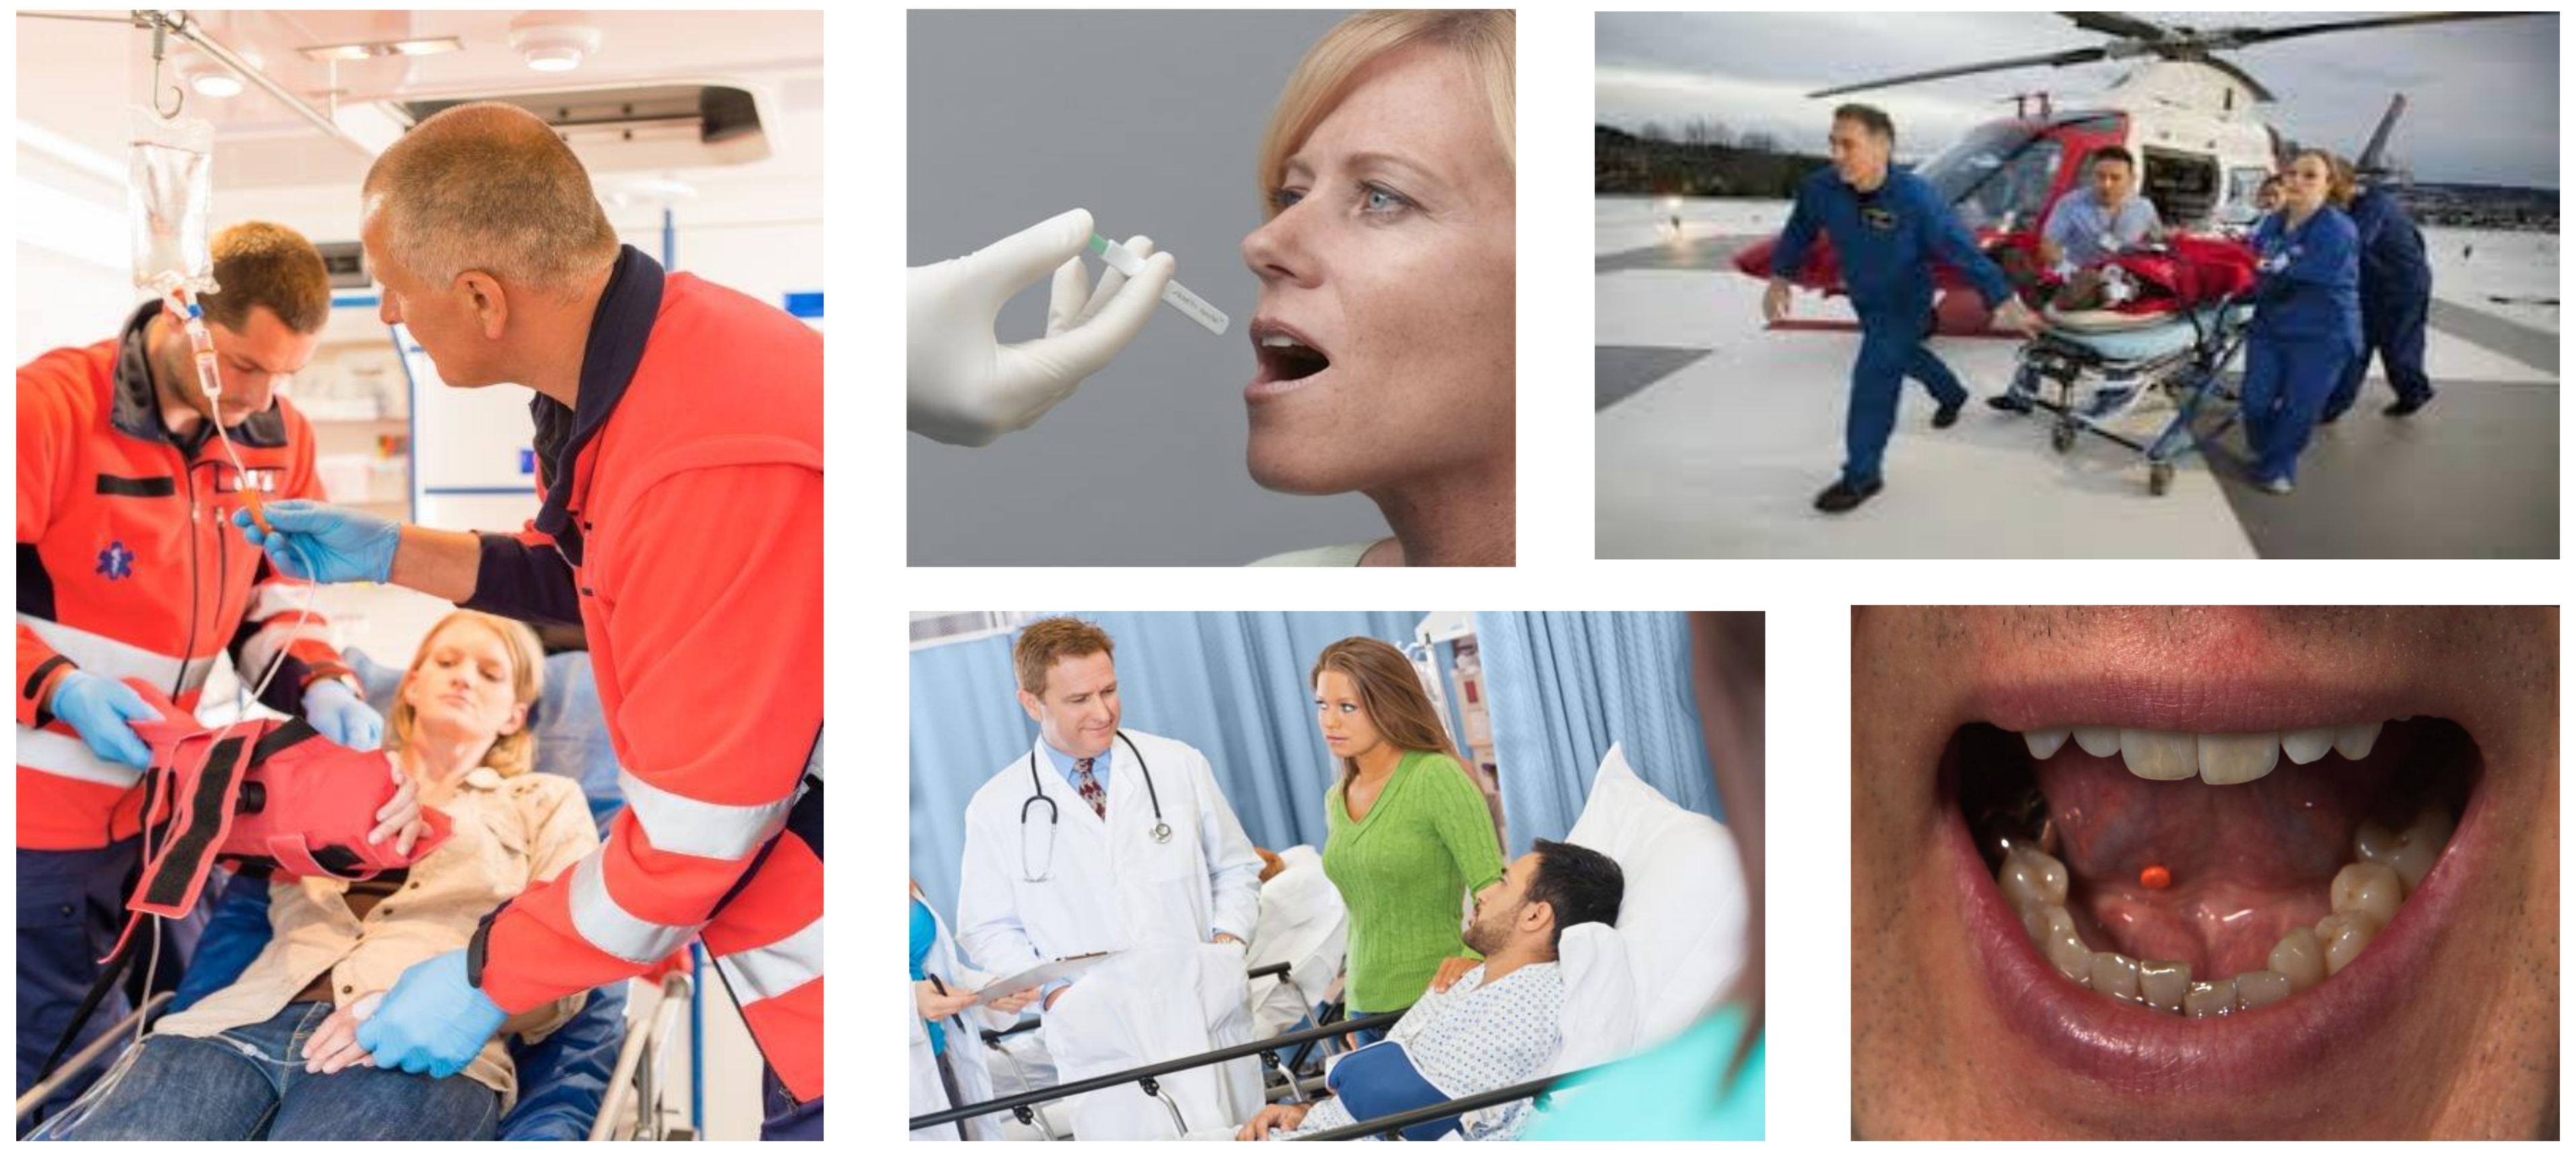

Methods

Study Procedures

- The study was a single-center, randomized, open-label, 2-sequence, 4-treatment, 4-period, crossover in healthy male and female adult subjects.
- Treatments were as follows:
 - **Treatment A**: Sufenta IV (50 mcg/mL) 30 mcg infused over 1 minute
 - **Treatment B**: Single-dose SST 30 mcg
 - **Treatment C**: 2 consecutive doses SST 15 mcg administered 20 minutes apart
 - **Treatment D**: 12 consecutive doses of SST 30 mcg administered 1 hour apart
- Subjects were enrolled and randomized to 1 of the 2 treatment sequences:
 - ABCD or ACBD;

Methods (Cont)

- Subjects received naltrexone orally to block the opioid effects of sufentanil;
- Treatment periods were separated by a 48-hour washout;
- Serial blood samples were collected pre-dose and up to 1440 minutes (24 hours) post dose;

Pharmacokinetic Parameters

The following key parameters were calculated from plasma concentration data of sufentanil:

- **$AUC_{0\text{-}inf}$** (h*pg/mL): Area under the plasma concentration-time curve from zero to infinity;
- **C_{max}** (pg/mL): Maximum observed plasma concentration;
- **T_{max}** (h): Time to reach the maximum plasma concentration;
- **$t_{1/2}$** (h): Apparent terminal elimination half-life;
- **$CST_{1/2}$** (h): Context-sensitive half-time measured as the time from T_{max} to time for plasma concentration to reach half of C_{max} following discontinuation of drug administration;
- **Bioavailability value** (F value) (%): The absolute amount of sufentanil absorbed.

Safety

- Subjects had screening and end-of-study Physical Exams, clinical laboratory tests (chemistry, hematology, and urinalysis), and ECGs;
- Safety was monitored via measurement of vital signs (including BP, HR, RR, temperature, and oxygen saturation) as well as assessment of Adverse Events (AE).

Table 1. Analysis of Key Sufentanil Pharmacokinetics (IV 30 mcg vs SST 30 mcg)

Pharmacokinetic Parameter	Treatment: (A) Sufenta IV 30 mcg (n=35)	Treatment: (B) Single SST 30 mcg (n=35)
$AUC_{0\text{-}inf}$ (h*pg/mL)		
Mean (SD)	539.7 (113.1)	277.7 (84.4)
Median	519.7	282.5
(Min, Max)	(387.0, 787.8)	(90.0, 451.2)
C_{max} (pg/mL)		
Mean (SD)	1073.9 (968.2)	63.14 (23.5)
Median	745.0	61.5
(Min, Max)	(201.0, 4510.0)	(21.0, 118.0)
T_{max} (hour)		
Mean (SD)	0.05 (0.03)	0.95 (0.33)
Median	0.07	1.00
(Min, Max)	(0.02, 0.17)	(0.50, 2.00)
$CST_{1/2}$ (hour)		
Mean (SD)	0.2 (0.2)	2.5 (0.9)
Median	0.1	2.3
(Min, Max)	(0.0, 0.7)	(0.8, 4.8)
Bioavailability (%)		
Mean (SD)	100	52.9

Results

Demographics

- Forty subjects (17 male, 23 female) were enrolled;
- Mean age was 30.5 years; mean BMI was 24.7 kg/m^2 (range: 18.7-29.6 kg/m^2);
- Of the 40 enrolled subjects, 35 (87.5%) were included in the PK analyses for Treatments A and B.

Primary Efficacy Variables

- Systemic exposure for sufentanil was higher following administration of Sufenta IV 30 mcg compared to single-dose SST 30 mcg;
 - Cmax was approximately 17 times higher via the IV route vs sublingual;
- Median $CST_{1/2}$ after IV administration was 0.10 hours (6 minutes) compared to 2.33 hours after administration of SST 30 mcg;
- Mean F Value (bioavailability) for the sublingual route was estimated at 53%; range was 20%-95%.

Safety

- In general, SST 30 mcg was safe and well tolerated when administered as single IV or sublingual dose with all but one treatment emergent AEs reported as mild
 - No deaths or serious AEs were reported during the study;
 - Two subjects discontinued due to treatment-related AEs of nausea (mild) and lip swelling (moderate; after administration of Treatment C)
- Twelve of 40 subjects (30.0%) reported a total of 23 AEs after administration of Sufenta IV. The most frequently reported AEs were gastrointestinal disorders (9 subjects [22.5%]), including nausea (5 subjects [12.5%]), and nervous system disorders (5 subjects [12.5%]), including dizziness (5 subjects [12.5%]).
- Nine of 39 subjects (23.1%) reported a total of 10 AEs after administration of single-dose SST 30 mcg. The most frequently reported were gastrointestinal disorders (7 subjects [17.9%]), including dyspepsia (3 subjects [7.7%]), and psychiatric disorders (2 subjects [5.1%]), including euphoric mood (2 subjects [5.1%]).

Conclusion

- **Systemic exposure of sufentanil after Sufenta IV 30 mcg was greater than after single-dose SST 30 mcg.**
- **Mean C_{max} was reduced by 17-fold and median $CST_{1/2}$ was extended by > 20-fold when Sufenta IV 30 mcg was compared to SST 30 mcg.**
- **In general, this study suggested sufentanil was safe and well tolerated when administered as single IV or sublingual dose.**

References

1. Sufenta® (Sufentanil Citrate Injection, USP) Package Insert, Akorn Inc. 2014
2. Melson TI et al. Sufentanil sublingual tablet System vs. intravenous patient-controlled analgesia with morphine for postoperative pain control: a randomized, active-comparator trial. Pain Pract 2014;14:679–88.
3. Minkowitz HS et al Sufentanil Sublingual Tablet 30 mcg for the Management of Pain Following Abdominal Surgery: A Randomized, Placebo-Controlled, Phase-3 Study. Pain Pract. 2016;epub ahead of print.
4. Maganti L, Panebianco DL, Maes AL. Evaluation of methods for estimating time to steady state with examples from Phase 1 studies. The AAPS Journal. March 2008;10(1):141-147

Poster presentation at the American Academy of Pain Medicine Meeting, Orlando, FL; March 16-19, 2017